2025 ESMO Asia *#818TiP*

Trial in progress: A Pivotal Phase 2, Randomized, Controlled, Open-Label, Trial (MIZAR-003) to Assess the Efficacy and Safety of GC101 TIL, an Autologous Tumor-Infiltrating Lymphocyte (TIL) Cell Therapy, for Patients with Advanced Anti-PD-1 Refractory Melanoma in Later-Line Therapy

Junjie Gu[1], Lu Si[2], Weizhen Zhang[3], Yu Chen[4], Jing Chen[5], Xiaoshi Zhang[6], Meiyu Fang[7], Dongyuan Zhu[8], Zhengyun Zou[9], Zhiguo Luo[10], Quanli Gao[11], Zhenchao Yuan[12], Di Wu[13], Jiwei Liu[14], Wang Ma[15], Xingxiang Pu[16], Huijing Feng[17], Jiyan Liu[18], Huajun Jin[19], Jun Guo[2]

1, Department of Melanoma and sarcoma, Peking University Cancer Hospital and Institute, Beijing, China; 2, Department of Melanoma and Sarcoma, Key Laboratory of Carcinogenesis and Translational Research (Ministry of Education/Beijing), Peking University Cancer Hospital and Institute, Beijing, China; 3, Medical Oncology, The Third People's Hospital of Zhengzhou, Zhengzhou, China; 4, Department of Hepatobiliary-Pancreatic Medical Oncology, Fujian Cancer Hospital and Fujian Medical University Cancer Hospital, Fuzhou, China; 5, Department of Oncology, Union Hospital, Tongji Medical College, Huazhong University of Science and Technology/ Cancer Center Union Hospital, Wuhan, China; 6, Department of Biological Therapy Center, Sun Yat-sen University Cancer Center, Guangzhou, China; 7, Key Laboratory of Head and Neck Oncology Rare diseases/head and neck medicine, Zhejiang Cancer Hospital - Cancer Research Institute, Hangzhou, China; 8, Rare Tumors Department, Shandong Cancer Hospital and Institute, Jinan, China; 9, The Comprehensive Cancer Center, The Affiliated Drum Tower Hospital of Nanjing University Medical School, Nanjing, China; 10, Medical Oncology Department, Fudan University Shanghai Cancer Center, Shanghai, China; 11, Department of Immunotherapy, Henan Cancer Hospital/Affiliated Cancer Hospital of Zhengzhou University, Zhengzhou, China; 12, Department of Bone and Soft Tissue Surgery, Affiliated Tumor Hospital of Guangxi Medical University, Nanning, China; 13, Cancer Centre, The First Hospital of Jilin University, Changchun, China; 14, Department of Oncology, The First Affiliated Hospital of Dalian Medical University, Dalian, China; 15, Department of Oncology, The First Affiliated Hospital of Zhengzhou University, Zhengzhou, China; 16, Thoracic Medicine Department, Hunan Cancer Hospital, Changsha, China; 17, Thoracic Oncology Department, Bethune Hospital, Taiyuan, China; 18, Department of Biotherapy and Cancer Center, State Key Laboratory of Biotherapy, West China School of Medicine/West China Hospital of Sichuan University, Chengdu, China; 19, Department of Medicine, Shanghai Juncell Therapeutics Co., Ltd, Shanghai, China.

Background

- Immune checkpoint inhibitors (ICIs) and targeted therapies have transformed the treatment landscape for advanced (unresectable or metastatic) melanoma. Nevertheless, most patients receiving frontline ICIs experience disease progression within one year [1-3].
- Furthermore, 40%–65% of patients exhibit primary resistance to ICIs, while 30%–40% develop secondary resistance [4-6].
- Autologous tumor-infiltrating lymphocyte (TIL) therapy has demonstrated clinically meaningful efficacy in patients with advanced melanoma following ICI treatment [7,8].
- However, the application of TIL therapy in advanced melanoma has been limited by the need for intensive lymphodepletion, using high-dose cyclophosphamide (CY) and fludarabine, along with high-dose interleukin-2 (IL-2), which are associated with significant toxicity.
- We explored GC101 TIL with a low-intensity preconditioning regimen, consisting of CY and hydroxychloroquine (HCQ), combined with IL-2–free TIL infusion. This regimen showed promising outcomes in a prior phase 1 trial [9].
- MIZAR-003 (NCT04943913) is a pivotal phase 2, multicenter, randomized, open-label, parallel-group, treatment study to assess the efficacy and safety of GC101 TIL therapy in melanoma patient refractory to programmed cell death protein 1(PD-1) antibodies.

Trial Design

Figure 1. MIZAR-003 Study Design

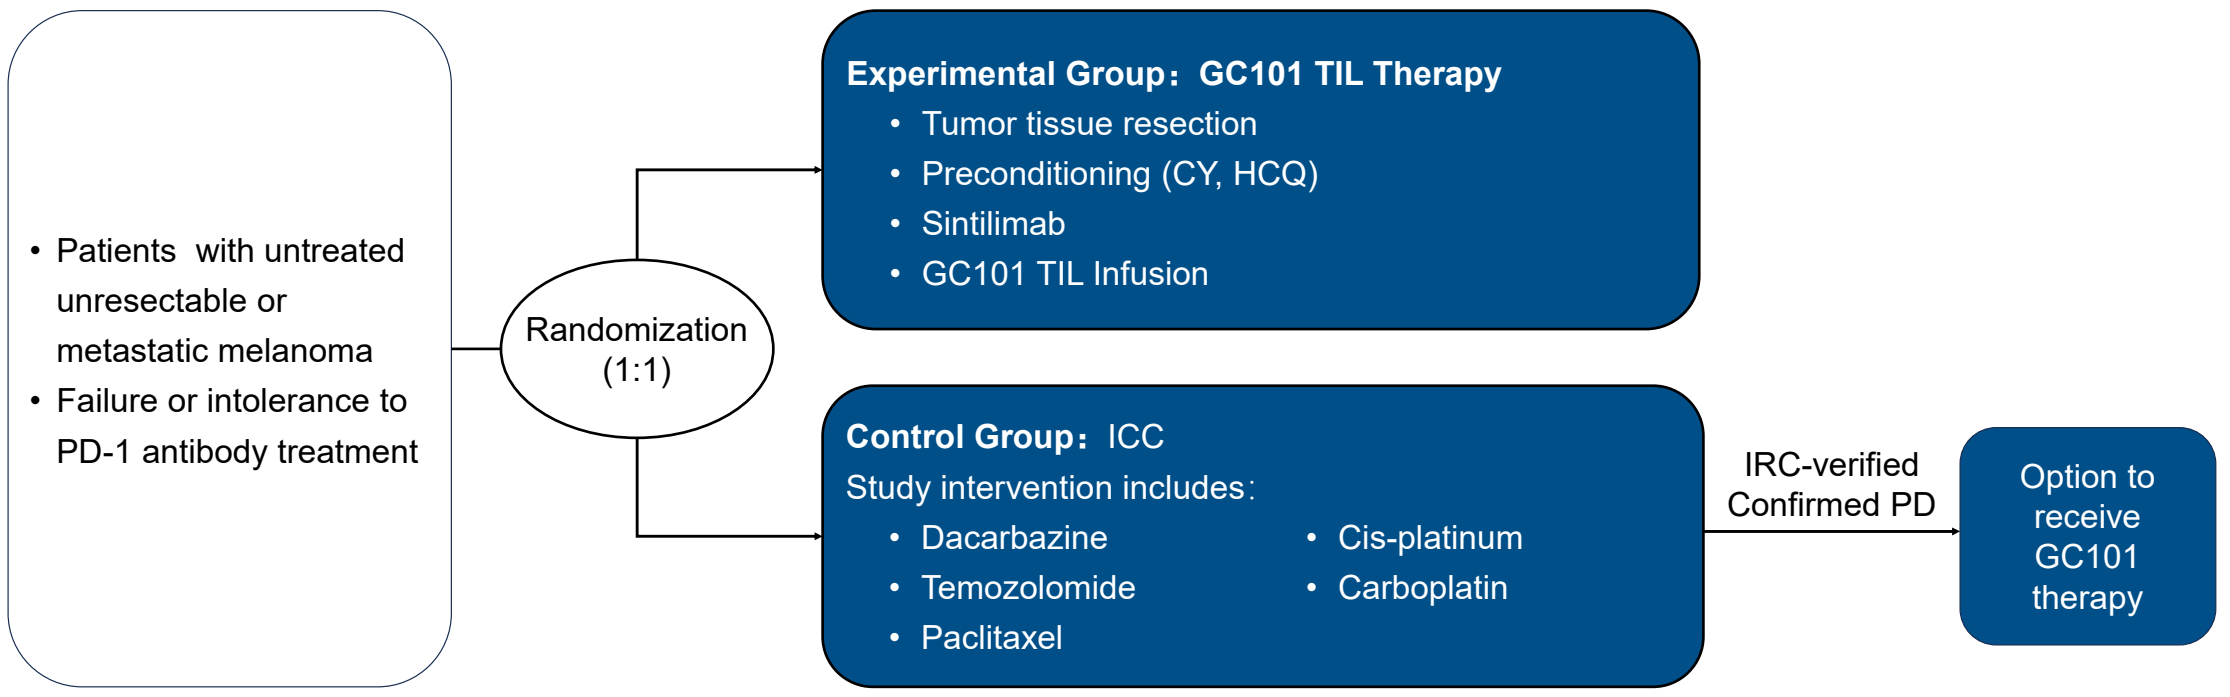
• Patients with untreated unresectable or metastatic melanoma
• Failure or intolerance to PD-1 antibody treatment
Randomization (1:1)
Experimental Group：GC101 TIL Therapy
• Tumor tissue resection
• Preconditioning (CY, HCQ)
• Sintilimab
• GC101 TIL Infusion
Control Group：ICC
Study intervention includes：
• Dacarbazine
• Temozolomide
• Paclitaxel
• Cis-platinum
• Carboplatin
IRC-verified Confirmed PD
Option to receive GC101 therapy

Study Endpoints

Primary endpoint:
- PFS as assessed by IRC per RECIST v1.1

Secondary endpoints:
- OS, PFS as assessed by the investigators per RECIST v1.1;
- PFS as assessed by the IRC and the investigators per iRECIST;
- ORR as assessed by the IRC and the investigators per RECIST v1.1 and iRECIST;
- DCR as assessed by the IRC and the investigators per RECIST v1.1 and iRECIST;
- DoR as assessed by the IRC and the investigators per RECIST v1.1 and iRECIST;
- Safety endpoints: Incidence of adverse events (AE) and serious adverse events (SAE)

Figure 2. MIZAR-003 Treatment Schema

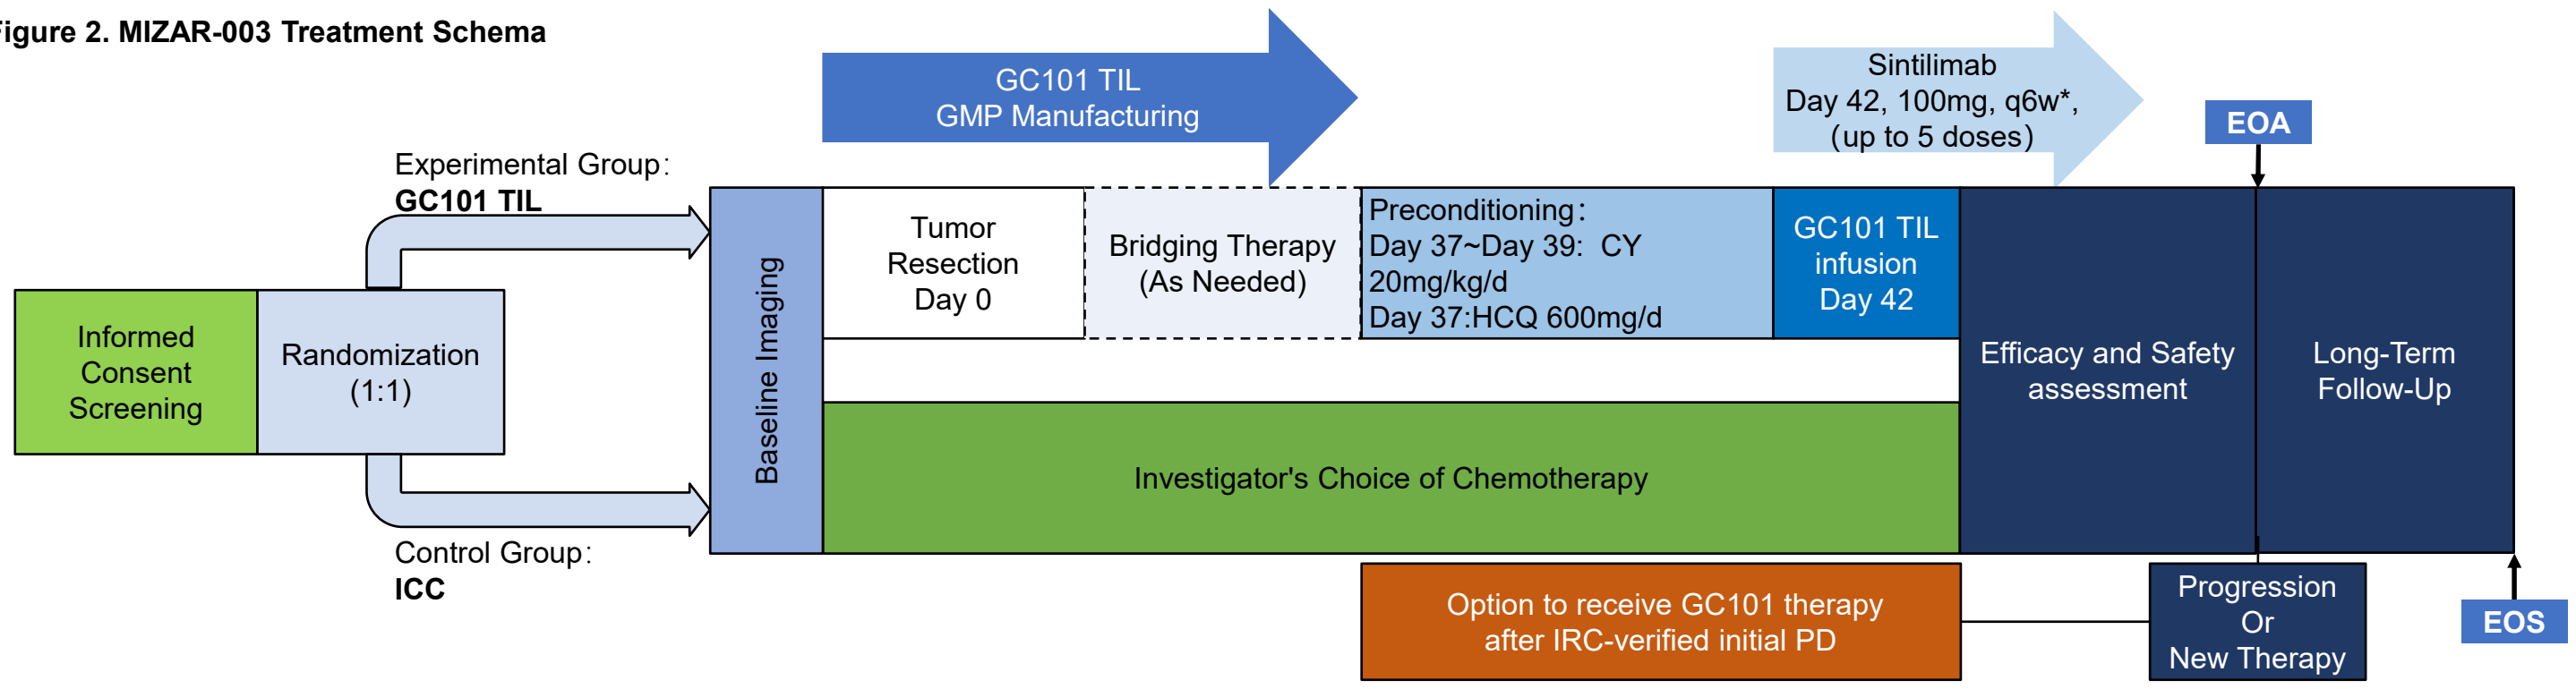
GC101 TIL GMP Manufacturing
Sintilimab Day 42, 100mg, q6w*, (up to 5 doses)
EOA
Experimental Group：GC101 TIL
Informed Consent Screening
Randomization (1:1)
Control Group：ICC
Baseline Imaging
Tumor Resection Day 0
Bridging Therapy (As Needed)
Preconditioning：Day 37~Day 39: CY 20mg/kg/d Day 37:HCQ 600mg/d
GC101 TIL infusion Day 42
Efficacy and Safety assessment
Long-Term Follow-Up
Investigator's Choice of Chemotherapy
Option to receive GC101 therapy after IRC-verified initial PD
Progression Or New Therapy
EOS

Key Eligibility Criteria

Inclusion Criteria
1. Age ≥ 18 years and ≤ 75 years, gender not restricted;
2. Patients with unresectable advanced, recurrent or metastatic melanoma (excluding uveal melanoma);
3. At least one of the following has occurred: failure of PD-1 antibody treatment or intolerance (if carrying BRAF V600 mutation, failure of BRAF and MEK inhibitors treatment; if carrying NRAS mutation, failure of toremifene treatment);
4. Have a feasible tissue region for tumor resection/puncture to isolate TILs;
5. According to RECIST 1.1 criteria, at least one measurable lesion remain after tissue sampling and tumor resection;
6. ECOG score 0-1;
7. Expected survival period greater than 3 months.

Exclusion Criteria
1. Melanoma of uveal/ocular origin;
2. Participated in other clinical trials of drug or biological therapy within the past 4 weeks;
3. Two or more types of malignant tumors;
4. Have signed the informed consent form or plan to receive attenuated live vaccine during the study;
5. Have not recovered to ≤ Grade 1 according to the National Cancer Institute - Common Toxicity Criteria Version 5.0 (NCI-CTCAE 5.0) (except for toxicities such as alopecia judged by the investigator to be without safety risks);
6. Any form of primary or acquired immunodeficiency.

References

1. Robert C, et al. Lancet Oncol. 2019;20(9):1239-1251.
2. Wolchok JD, et al. J Clin Oncol. 2022;40(2):127-137.
3. Tawbi HA, et al. N Engl J Med. 2022;386(1):24-34.
4. Mooradian MJ, et al. Oncology (Williston Park). 2019;33(4):141-148.
5. Hamid O, et al. Ann Oncol. 2019;30(4):582-588.
6. Wolchok JD, et al. N Engl J Med. 2017;377(14):1345-1356.
7. Sarnaik A, et al. Presented at SITC 2022.
8. Chesney J, et al. J Immunother Cancer. 2022;10: e005755.
9. Guo J., et al. BMC Med. 2024; 22(1): p. 207.

Abbreviations

AE, adverse event; CY, cyclophosphamide; DCR, disease control rate; DoR, duration of response; ECOG, Eastern Cooperative Oncology Group; EOA, end of assessment; EOS, end of study; EOT, end of treatment; GMP, good manufacturing practice; HCQ, hydroxychloroquine; ICC, investigator's choice of chemotherapy; ICI, immune checkpoint inhibitor; IL-2, interleukin-2; IRC, independent review committee; NCI-CTCAE, national cancer institute common terminology criteria for adverse events; ORR, objective response rate; OS, overall survival; PD, progressive disease; PD-1, programmed cell death protein 1; PFS, progression-free survival; SAE, serious adverse event; TIL, tumor infiltrating lymphocyte.

Acknowledgments:

•The authors would like to the investigators and study site team members who are participating in the study.

•This study is sponsored by Shanghai Juncell Therapeutics Co., Ltd.